Effect of Secukinumab Versus Adalimumab Biosimilar on Radiographic Progression in Patients With Radiographic Axial Spondyloarthritis: A Randomized Phase IIIb Study

L15

Xenofon Baraliakos[1], Mikkel Østergaard[2], Denis Poddubnyy[3], Désirée van der Heijde[4], Atul Deodhar[5], Pedro M. Machado[6], Victoria Navarro-Compán[7], Kay Geert Hermann[8], Mitsumasa Kishimoto[9], Eun Young Lee[10], Lianne S. Gensler[11], Uta Kiltz[1], Marco F. Eigenmann[12], Patricia Pertel[12], Aimee Readie[13], Hanno B. Richards[12], Brian Porter[13], Juergen Braun[1]

[1]Rheumazentrum Ruhrgebiet, Herne, Ruhr-Universität Bochum, Bochum, Germany; [2]Center for Rheumatology and Spine Diseases, Center of Head and Orthopedics, Copenhagen University Hospital Rigshospitalet, Glostrup, Denmark; [3]Charité - Universitätsmedizin Berlin and German Rheumatism Research Centre, Berlin, Germany; [4]Department of Rheumatology, Leiden University Medical Center, Leiden, The Netherlands; [5]Division of Arthritis and Rheumatic Diseases, Oregon Health & Science University, Portland, USA; [6]Centre for Rheumatology & Department of Neuromuscular Diseases, University College London, London, UK; [7]Department of Rheumatology, University Hospital La Paz, IdiPaz, Madrid, Spain; [8]Department of Radiology, University Hospital Charité - Campus Mitte, Berlin, Germany; [9]Department of Nephrology and Rheumatology, Kyorin University School of Medicine, Tokyo, Japan; [10]Division of Rheumatology, Department of Internal Medicine, Seoul National University College of Medicine, Seoul, South Korea; [11]Department of Medicine, Division of Rheumatology, University of California, San Francisco, California, USA; [12]Novartis Pharma AG, Basel, Switzerland; [13]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA

INTRODUCTION

- Biological disease-modifying antirheumatic drugs (bDMARDs) are clinically efficacious in patients with axial spondyloarthritis (axSpA) including radiographic axSpA (r-axSpA)[1]
- In addition to improving symptoms and function, an important goal of management of axSpA is preventing potentially irreversible structural damage[2]
- Limited data exist on the effect of bDMARDs in slowing radiographic progression in patients with r-axSpA. Two-year data from MEASURE 1 showed low radiographic progression with secukinumab[3]
- Here, we report data from SURPASS,[4] the first head-to-head study in patients with r-axSpA, that compared the effect of secukinumab versus adalimumab biosimilar (SDZ-ADL) on spinal radiographic progression

METHODS

Study Design and Patients

- The study methodology has been described in detail previously.[4] SURPASS was a phase IIIb study that enrolled biologic-naïve patients with active r-axSpA (i.e., ankylosing spondylitis [AS]) with a Bath Ankylosing Spondylitis Disease Activity Index (BASDAI) ≥4, spinal pain score ≥4 (range 0–10), and total back pain score ≥40 mm (range 0–100 mm)
- Patients were also required to have high-sensitivity C-reactive protein (hs-CRP) ≥5 mg/L or ≥1 syndesmophyte(s) on spinal radiograph (predictors of radiographic progression)
- Eligible patients were randomized (1:1:1) to dose-blinded secukinumab 150 mg or 300 mg, or open-label SDZ-ADL 40 mg
- All radiographs and MRIs were reviewed by 3 independent central readers blinded to treatment arm and chronology of images

Assessments

- Primary endpoint was the proportion of patients with no radiographic progression (change from baseline [CFB] in modified Stoke AS Spinal Score [mSASSS] ≤0.5; mean score of 3 readers) on secukinumab versus SDZ-ADL at week 104 (superiority testing)[4]
- Key secondary endpoints included CFB-mSASSS by week 104 and proportion of patients with ≥1 syndesmophyte(s) at baseline with no new syndesmophyte(s) at week 104
- Other secondary endpoints included CFB-MRI Berlin sacroiliac joint (SIJ) edema score, CFB-AS Spine MRI-activity (ASspiMRI-a) Berlin modification score, and safety

RESULTS

Demographic and Baseline Disease Characteristics

- Overall, 859 patients received secukinumab 150 mg (N=287), 300 mg (N=286), or SDZ-ADL 40 mg (N=286)
- Patient demographics and baseline disease characteristics were balanced across treatment arms. Baseline parameters indicated that the population was at high risk of radiographic progression (e.g., increased CRP levels, presence of syndesmophytes, smokers; **Table 1**)

Table 1. Demographic and baseline disease characteristics

Characteristics, mean (SD) unless specified otherwise	SEC 150 mg N=287	SEC 300 mg N=286	SDZ-ADL 40 mg N=286	Total N=859
Age, years	42.1 (12.0)	42.2 (12.5)	41.9 (12.7)	42.1 (12.4)
Male, n (%)	230 (80.1)	223 (78.0)	221 (77.3)	674 (78.5)
BMI, kg/m^2	27.7 (5.7)	26.9 (5.5)	27.2 (5.4)	27.3 (5.5)
Smoking status, n (%)				
Former	58 (20.2)	54 (18.9)	45 (15.7)	157 (18.3)
Current	85 (29.6)	82 (28.7)	80 (28.0)	247 (28.8)
Time since first diagnosis for AS, years	6.4 (9.0)	6.6 (8.4)	7.1 (10.1)	6.7 (9.2)
mSASSS (0-72)	17.6 (21.3)	16.5 (20.8)	15.7 (19.5)	16.6 (20.6)
Patients with syndesmophytes, n (%)	211 (73.5)	204 (71.3)	212 (74.1)	627 (73.0)
Number of syndesmophytes	7.3 (7.8)	7.0 (7.6)	6.7 (7.3)	7.0 (7.6)
Total back pain (0-100 mm)	72.6 (15.9)	73.0 (16.0)	72.7 (16.8)	72.8 (16.3)
BASFI (0-10)	6.7 (1.9)	6.7 (2.0)	6.5 (2.1)	6.6 (2.0)
BASDAI (0-10)	7.1 (1.4)	7.2 (1.4)	7.2 (1.5)	7.1 (1.4)
hsCRP (mg/L)	20.8 (28.6)	20.7 (26.5)	19.8 (22.6)	20.4 (26.0)
HLA-B27 positive, n (%)	235 (81.9)	227 (79.4)	236 (82.5)	698 (81.3)

AS, ankylosing spondylitis; BASDAI, Bath Ankylosing Spondylitis Disease Activity Index; BASFI, Bath Ankylosing Spondylitis Functional Index; BMI, body mass index; HLA-B27, Human leukocyte antigen B27; hsCRP, high sensitivity C-reactive protein; mSASSS, modified Stoke Ankylosing Spondylitis Spinal Score; SD, standard deviation; SDZ-ADL, adalimumab biosimilar; SEC, secukinumab

Efficacy

Cumulative probability of change from baseline in mSASSS

- As seen in **Figure 1**, the cumulative distribution of CFB-mSASSS was similar across treatment arms

Figure 1. Probability plot of change from baseline in mSASSS at week 104

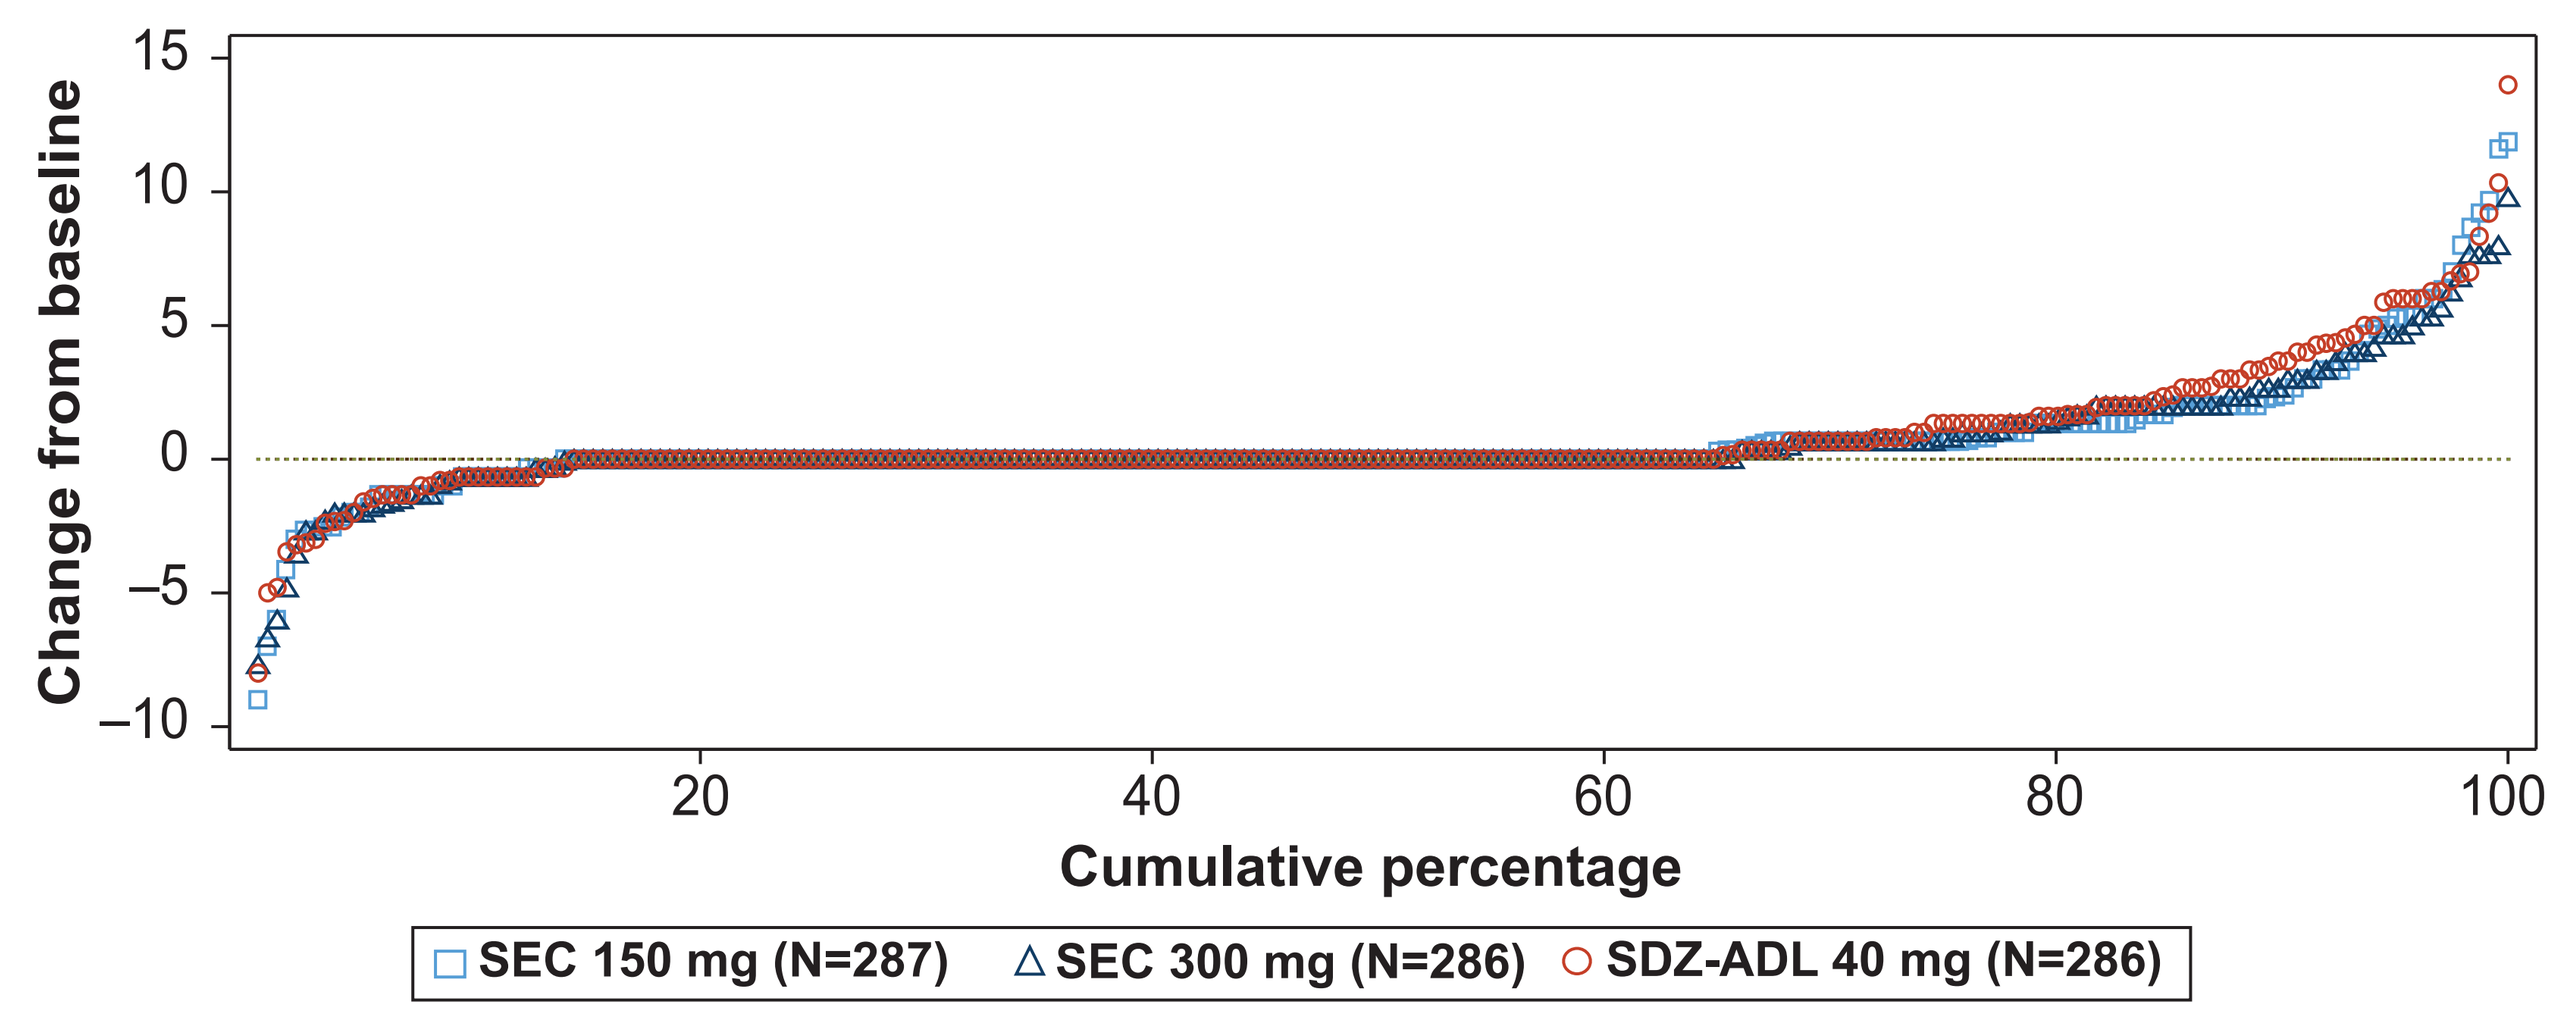
Change from baseline
15
10
5
0
−5
−10
20
40
60
80
100
Cumulative percentage
□ SEC 150 mg (N=287) △ SEC 300 mg (N=286) ○ SDZ-ADL 40 mg (N=286)

Each point represents an individual patient's change from baseline over 2 years
mSASSS, modified Stoke Ankylosing Spondylitis Spinal Score; SDZ-ADL, adalimumab biosimilar; SEC, secukinumab

Primary endpoint

- At week 104, the proportion of patients with no radiographic progression (CFB-mSASSS ≤0.5) was 66.1%, 66.9%, and 65.6% in the secukinumab 150 mg, 300 mg, and SDZ-ADL arms, respectively (**Table 2**); *P*=not significant for both secukinumab doses versus SDZ-ADL. Statistical testing was stopped beyond the primary endpoint hypothesis

Table 2. Proportion of patients with no radiographic progression (mSASSS progression of ≤0.5) at week 104

Treatment Group	n	No Progression Rate (%)	Estimated Mean (95% CI)	Marginal Difference (95% CI) vs SDZ-ADL 40 mg	Nominal *P* value
SEC 150 mg (N=287)	283	66.1	66.63 (60.73 to 72.54)	1.51 (−6.63 to 9.64)	0.716
SEC 300 mg (N=286)	280	66.9	66.80 (60.45 to 73.14)	1.67 (−6.61 to 9.95)	0.693
SDZ-ADL 40 mg (N=286)	283	65.6	65.13 (58.77 to 71.49)	-	-

mSASSS scores are based on the average score of 3 readers. No adjudication was performed.
Missing responses at week 104 were multiply imputed. Estimated mean, marginal difference, 95% CI, and *P* values are from a logistic regression model with treatment as a factor and baseline mSASSS score as a covariate using marginal standardization method.
CI, confidence interval; mSASSS, modified Stoke Ankylosing Spondylitis Spinal Score; n, number of patients with measures at baseline visit; SDZ-ADL, adalimumab biosimilar; SEC, secukinumab

Key secondary endpoints

- Mean CFB-mSASSS at week 104 was 0.54, 0.55, and 0.72 in the secukinumab 150 mg, 300 mg, and SDZ-ADL arms, respectively (**Table 3**)

Table 3. Change from baseline in mSASSS at week 104

Treatment Group	n	Within Treatment	Treatment Contrast in LS Mean vs SDZ-ADL 40 mg	
		LS Mean (SE)	LS Mean (SE)	95% CI
SEC 150 mg (N=287)	283	0.54 (0.18)	−0.18 (0.24)	−0.65 to 0.29
SEC 300 mg (N=286)	280	0.55 (0.18)	−0.16 (0.24)	−0.64 to 0.32
SDZ-ADL 40 mg (N=286)	283	0.72 (0.18)	-	-

mSASSS scores are based on the average score of 3 readers. No adjudication was performed.
Missing mSASSS values at week 104 were multiply imputed. LS mean and 95% CI are from an ANCOVA model with treatment as a factor and baseline mSASSS score as a covariate.
CI, confidence interval; LS, least square; mSASSS, modified Stoke Ankylosing Spondylitis Spinal Score; n, number of patients with measures at baseline visit; SDZ-ADL, adalimumab biosimilar; SE, standard error; SEC, secukinumab

- Overall, 56.9%, 53.8%, and 53.3% of patients with ≥1 syndesmophyte(s) at baseline in the secukinumab 150 mg, 300 mg, and SDZ-ADL arms, respectively, did not develop new syndesmophyte(s) by week 104 (**Table 4**)

Table 4. Percentage of patients with syndesmophyte(s) at baseline and no new syndesmophyte(s) at week 104

Treatment Group	Patients With Syndesmophyte(s) at Baseline, n (%)	Patients With No New Syndesmophyte(s) (%)	Estimated Mean (95% CI)	Marginal Difference (95% CI) vs SDZ-ADL 40 mg
SEC 150 mg (N=287)	211 (73.5)	56.9	57.22 (50.16 to 64.28)	4.32 (−5.62 to 14.27)
SEC 300 mg (N=286)	204 (71.3)	53.8	53.98 (46.19 to 61.78)	1.09 (−9.13 to 11.31)
SDZ-ADL 40 mg (N=286)	212 (74.1)	53.3	52.89 (45.54 to 60.24)	-

A patient was considered to have a syndesmophyte if ≥1 reader assessed mSASSS score of ≥2 for any individual vertebral corner.
Missing responses at week 104 were multiply imputed. Estimated mean, marginal difference, and 95% CI are from a logistic regression model with treatment as a factor and baseline count of vertebral corners with syndesmophyte as a covariate using marginal standardization method.
CI, confidence interval; SDZ-ADL, adalimumab biosimilar; SEC, secukinumab

MRI

- **Figures 2a** and **2b** show the baseline and postbaseline scores at week 16 and week 104 for SIJ and spine, respectively, across the treatment groups

Figure 2. MRI observed data, N=418, a) SIJ edema by Berlin active inflammatory lesions scoring (0–24 scale), b) Spine edema by the ASspiMRI-a Berlin modification scoring (0–69 scale)

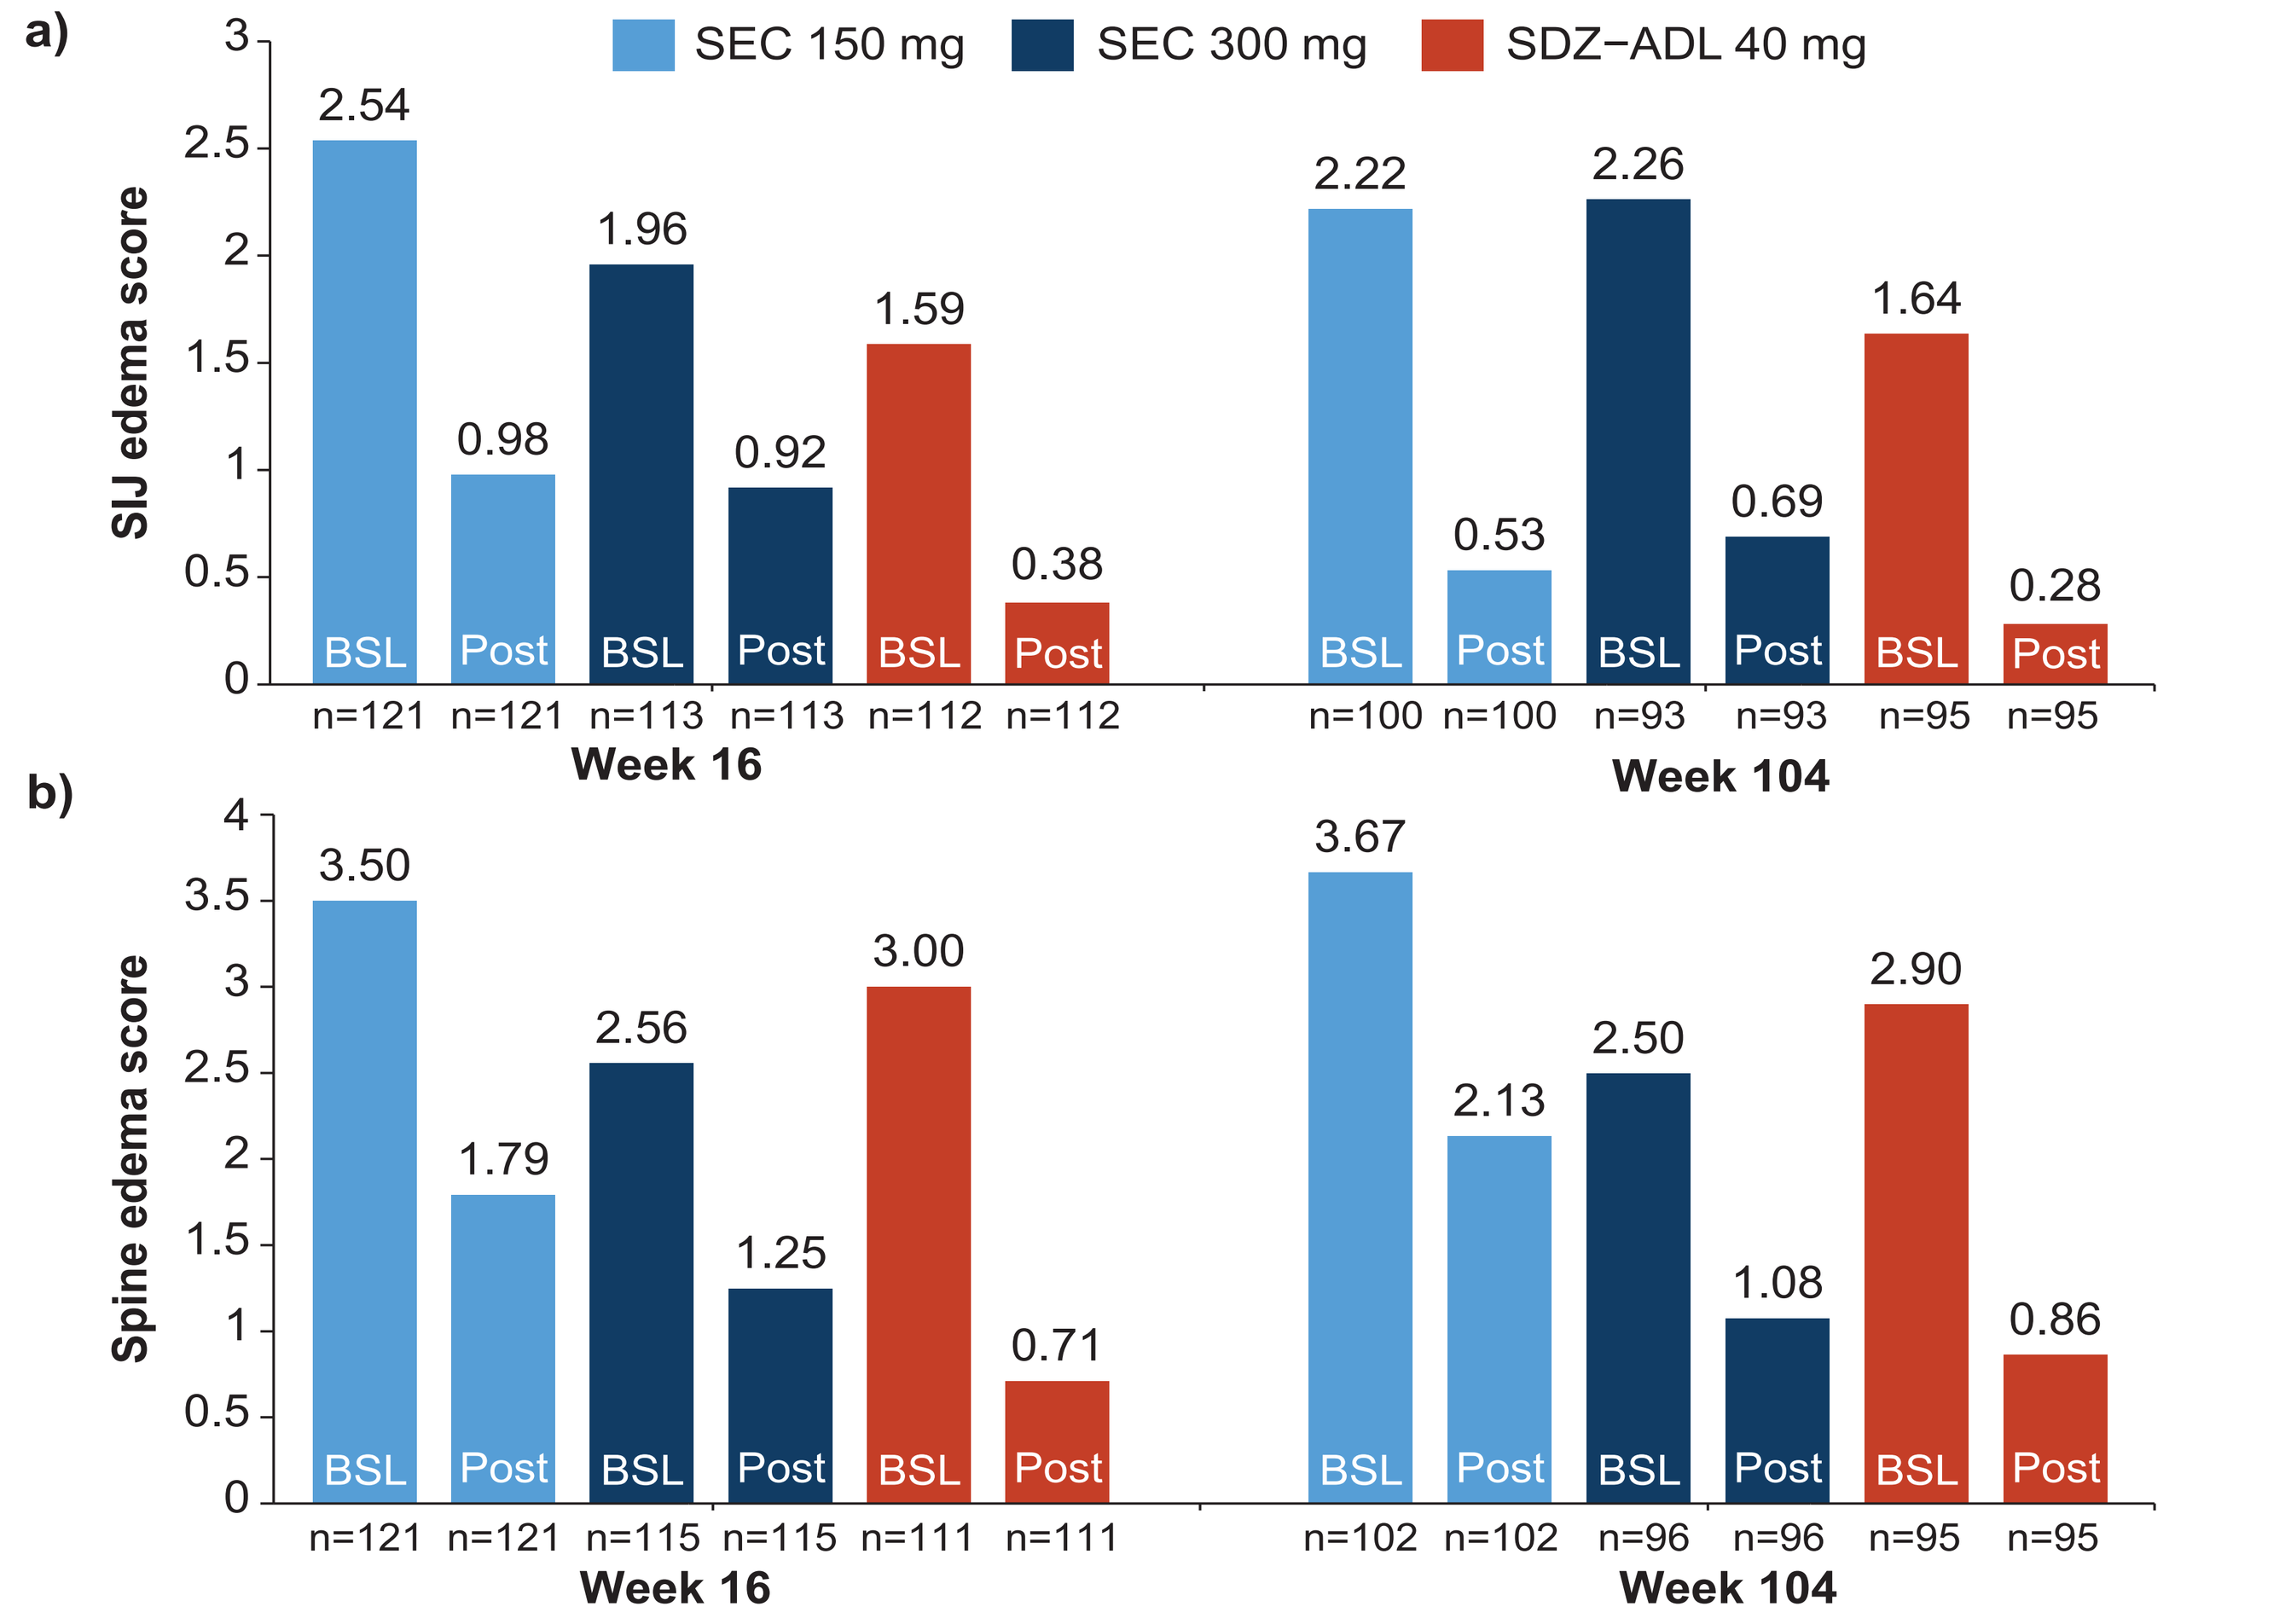
a)
SEC 150 mg
SEC 300 mg
SDZ−ADL 40 mg
SIJ edema score
2.54
0.98
1.96
0.92
1.59
0.38
2.22
0.53
2.26
0.69
1.64
0.28
BSL Post BSL Post BSL Post
n=121 n=121 n=113 n=113 n=112 n=112
Week 16
n=100 n=100 n=93 n=93 n=95 n=95
Week 104
b)
Spine edema score
3.50
1.79
2.56
1.25
3.00
0.71
3.67
2.13
2.50
1.08
2.90
0.86
n=121 n=121 n=115 n=115 n=111 n=111
Week 16
n=102 n=102 n=96 n=96 n=95 n=95
Week 104

Data are presented as observed.
BSL, baseline; n, number of patients at the specific time point; SDZ-ADL, adalimumab biosimilar; SEC, secukinumab; SIJ, sacroiliac joint

Safety

- Overall, 79.7%, 81.8%, and 84.2% of patients had ≥1 adverse event (AE), and 14.0%, 10.2%, and 11.2% of patients had ≥1 serious AE in the secukinumab 150 mg, 300 mg, and SDZ-ADL arms, respectively
- The most common AE was nasopharyngitis (16.4%, 14.0%, and 15.4% in the secukinumab 150 mg, 300 mg, and SDZ-ADL arms, respectively)
- The frequency of AEs was similar between secukinumab and SDZ-ADL arms, except for Crohn's disease (exposure-adjusted incidence rate per 100 patient-years [EAIR]: secukinumab 1.0; SDZ-ADL 0.2), ulcerative colitis (EAIR: secukinumab 0.2; SDZ-ADL 0.0), uveitis (EAIR: secukinumab 2.1; SDZ-ADL 1.4), and pulmonary tuberculosis (EAIR: secukinumab 0.0; SDZ-ADL 0.2)

CONCLUSION

- Spinal radiographic progression over 2 years was low with no significant difference between secukinumab and adalimumab biosimilar arms
- Safety was consistent with the well-established safety profiles of secukinumab and adalimumab biosimilar

REFERENCES

1. Sepriano A et al. *RMD Open.* 2017;3(1):e000396
2. van der Heijde D et al. *Ann Rheum Dis.* 2017;76(6):978-991
3. Braun J et al. *Ann Rheum Dis.* 2017;76(6):1070-1077
4. Baraliakos X et al. *Clin Drug Investig.* 2020;40(3):269-278

Acknowledgments

The authors thank Kshama Chitnis and Mohammad Fahad Haroon (Novartis, India) for medical writing support.
The study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at: American College of Rheumatology (ACR) Convergence 2022, Philadelphia, Pennsylvania, November 10–14, 2022

To download a copy of this poster, visit the web at: http://novartis.medicalcongressposters.com/Default.aspx?doc=09fa1

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without the written permission of the authors.

Scan this QR code to download the poster