Avacopan Versus Prednisone Taper in Patients With ANCA-Associated Vasculitis Without Kidney Involvement in a Phase 3 Trial

Christian Pagnoux[1], Antoine Néel[2], Sarah Bray[3], Rachel E. Gurlin[4], Darcy Trimpe[4], David R.W. Jayne[5], and Peter A. Merkel[6] for the ADVOCATE Study Group

[1]Mount Sinai Hospital, University of Toronto, Toronto, Ontario, Canada; [2]Internal Medicine, CHU Nantes, Nantes, France; [3]Amgen Ltd, Cambridge, UK; [4]Amgen Inc., Thousand Oaks, CA, USA; [5]University of Cambridge, Cambridge, UK; [6]University of Pennsylvania, Philadelphia, PA, USA

Introduction

- Anti-neutrophil cytoplasmic autoantibody (ANCA)-associated vasculitis (granulomatosis with polyangiitis [GPA] and microscopic polyangiitis [MPA]) frequently involves the kidneys[1]
- However, some patients with GPA or MPA have no kidney involvement, and studies in this group are limited

Objective

To report a post hoc subgroup analysis of 62 patients (32 in the avacopan group, 30 in the prednisone taper group) without active kidney involvement at baseline in the ADVOCATE trial

ADVOCATE Phase 3 Trial[2]

331 patients randomized

166 patients 1:1 **165 patients**

Avacopan
30 mg twice daily
+
Placebo (matching prednisone taper)

Placebo (matching avacopan)
+
Prednisone Taper
60 mg tapered to 0 mg over 20 weeks

NCT02994927; 52-week study

Key inclusion criteria:
- Newly diagnosed or relapsing GPA or MPA
- Proteinase-3 (PR3)-ANCA or myeloperoxidase (MPO)-ANCA positive (at enrollment or in the past)
- Birmingham Vasculitis Activity Score (BVAS): ≥ 1 major item, or ≥ 3 minor items, or at least both hematuria and proteinuria

Background therapy (all patients):
- Rituximab (RTX), cyclophosphamide (CYC)/azathioprine, or CYC/mycophenolate mofetil
- Non–study-supplied glucocorticoids (GCs) under protocol-specified conditions

Results

Baseline Characteristics

Baseline Characteristics	Avacopan (N = 32)	Prednisone Taper (N = 30)
Age (years), mean ± SD	62.4 ± 14.4	53.2 ± 15.2
Male / female, n (%)	14 (43.8) / 18 (56.3)	12 (40.0) / 18 (60.0)
BVAS, mean ± SD	11.7 ± 3.6	10.3 ± 4.0
Newly diagnosed / relapsed, n (%)	17 (53.1) / 15 (46.9)	14 (46.7) / 16 (53.3)
PR3-ANCA / MPO-ANCA, n (%)	19 (59.4) / 13 (40.6)	23 (76.7) / 7 (23.3)
GPA / MPA, n (%)	26 (81.3) / 6 (18.8)	27 (90.0) / 3 (10.0)
RTX IV / CYC IV or oral, n (%)	26 (81.3) / 6 (18.8)	25 (83.3) / 5 (16.7)
BVAS organ involvement, n (%)		
General	28 (87.5)	21 (70.0)
Ear, nose, throat	26 (81.3)	25 (83.3)
Lung	22 (68.8)	16 (53.3)
Mucous membranes/eyes	7 (21.9)	8 (26.7)
Cutaneous	2 (6.3)	6 (20.0)
Nervous system	7 (21.9)	3 (10.0)
Abdominal	0 (0.0)	1 (3.3)
Cardiovascular	3 (9.4)	0 (0.0)

Efficacy Outcomes

Remission at Week 26, % (n/N)

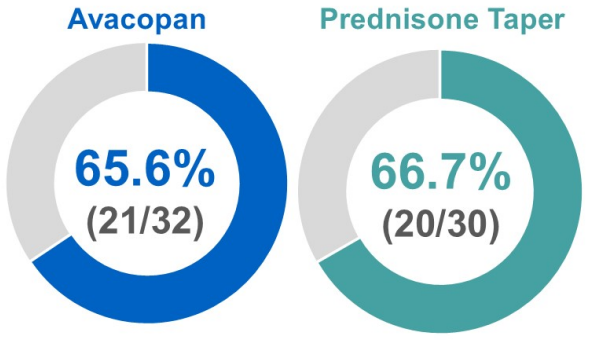
Avacopan
Prednisone Taper
65.6%
(21/32)
66.7%
(20/30)

Difference (two-sided 95% CI): –1.0% (–24.6, 22.5)

Sustained Remission at Week 52, % (n/N)

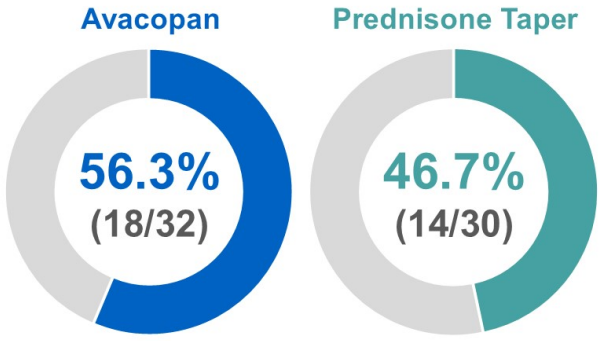
Avacopan
Prednisone Taper
56.3%
(18/32)
46.7%
(14/30)

Difference (two-sided 95% CI): 9.6% (–15.2, 34.4)

Relapse Rate After BVAS=0 at Any Time Was Achieved, n/N' (%)

Avacopan		Prednisone Taper
4/30 (13.3%)	vs	6/28 (21.4%)

Hazard ratio (95% CI): 0.58 (0.16, 2.06)

Glucocorticoid Doses During the Trial

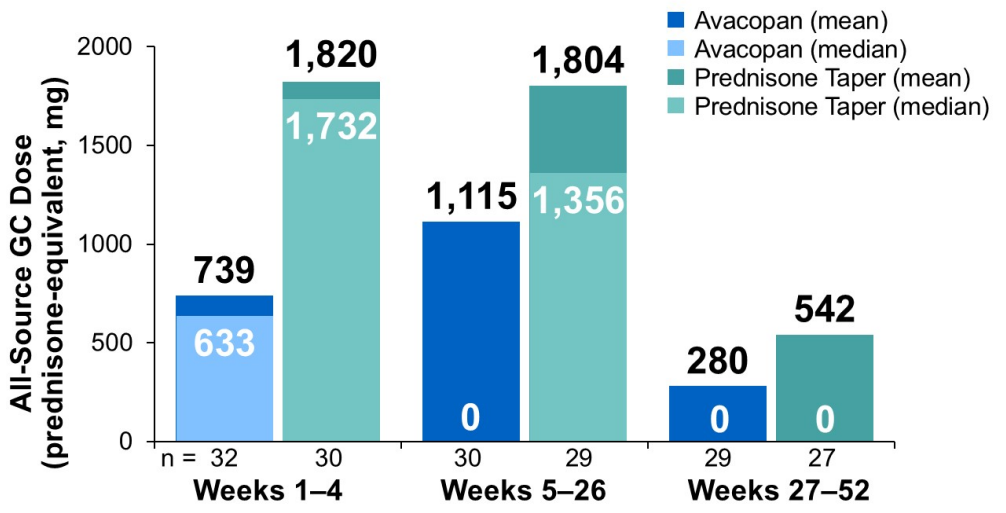
Avacopan (mean)
Avacopan (median)
Prednisone Taper (mean)
Prednisone Taper (median)
All-Source GC Dose (prednisone-equivalent, mg)
2000
1500
1000
500
0
739
633
1,820
1,732
1,115
0
1,804
1,356
280
0
542
0
n = 32
30
30
29
29
27
Weeks 1–4
Weeks 5–26
Weeks 27–52

GC Toxicity and Health-Related Quality of Life (HRQoL)

- Patients receiving avacopan experienced less GC-related toxicity than those receiving a prednisone taper
- HRQoL improvements were observed in patients treated with avacopan or a prednisone taper

Safety Outcomes

Events, No. of patients (%), No of events	Avacopan (N = 32)	Prednisone Taper (N = 30)
Any adverse events	32 (100.0) 314 events	28 (93.3) 362 events
Any serious adverse events	9 (28.1) 12 events	9 (30.0) 18 events
Deaths	0 (0)	1 (3.3)

More Information

Scan the QR code for further information on methods, results on Glucocorticoid Toxicity Index (GTI), and HRQoL, as well as disclosures, references, abbreviations and acknowledgments

Summary

Among patients with GPA or MPA without kidney involvement in the ADVOCATE trial, in combination with immunosuppression therapy, treatment with avacopan versus a prednisone taper was accompanied by numerically:
- **Higher sustained remission at week 52**
- **Lower relapse rate**
- **Lower glucocorticoid dose and toxicity**
- **Improvements in health-related quality of life**
- **Comparable safety profiles between treatment groups**

Efficacy and safety outcomes observed between treatment groups in this subgroup of patients were similar to those of the entire trial cohort[2]

CONTACT | medinfo@amgen.com

© 2024 Amgen Inc. All rights reserved