Poster **0589**

Laura Coates, MRCP, PhD | laura.coates@ndorms.ox.ac.uk

Time to Clinical Response to Secukinumab Across Disease Domains Among Patients With Psoriatic Arthritis: a Pooled Post Hoc Analysis of 4 Phase 3 Trials

Laura Coates, MRCP, PhD,[1] Iain McInnes, MD,[2] M. Elaine Husni, MD, MPH,[3] Cynthia Vizcaya, MD,[4] Weibin Bao, MsC,[5] Philip Mease, MD[6]

[1]Nuffield Department of Orthopaedics, Rheumatology and Musculoskeletal Sciences, University of Oxford, Oxford, UK; [2]College of Medical, Veterinary and Life Sciences, University of Glasgow, Glasgow, UK; [3]Department of Rheumatic and Immunologic Diseases, Cleveland Clinic, Cleveland, OH, USA; [4]Novartis Pharma AG, Basel, Switzerland; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]Swedish Medical Center/Providence St. Joseph Health and University of Washington, Seattle, WA, USA

KEY FINDINGS & CONCLUSIONS

- In this post hoc analysis, patients with PsA receiving secukinumab 300 or 150 mg in the FUTURE 2-5 trials demonstrated rapid and sustained clinical responses across key GRAPPA-OMERACT domains, including musculoskeletal disease activity, skin disease, and systemic inflammation
- MCID in SJC66, TJC68, enthesitis, and dactylitis was observed as early as 2 to 4 weeks, along with reductions in CRP levels, reflecting the anti-inflammatory effect of secukinumab
- Notably, the impact of secukinumab extended beyond clinical efficacy endpoints as patients with PsA experienced early and meaningful improvements in HRQOL, physical function, and pain. These improvements occurred in parallel with the resolution of musculoskeletal symptoms, highlighting the efficacy of secukinumab in both clinical and patient-reported outcomes
- Further exploration into the timing of domain-specific responses could offer new insights into optimal disease management strategies for patients with PsA

Scan to obtain:
- Poster

https://www.medicalcongressposters.com//Default.aspx?doc=ef8fb

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study was sponsored by Novartis Pharmaceuticals Corporation.
Poster presented at American College of Rheumatology Convergence 2024, November 14-19, 2024; Washington DC

BACKGROUND

- Psoriatic arthritis (PsA) is a progressive inflammatory disease that can manifest with considerable clinical heterogeneity across a range of disease domains, including skin and nail changes, peripheral joint inflammation or damage, enthesitis, dactylitis, and spondyloarthritis, either alone or in combination[1]
- The PsA core set of domains, updated in 2016 by the Group for Research and Assessment of Psoriasis and Psoriatic Arthritis (GRAPPA) and endorsed by the Outcome Measures in Rheumatology (OMERACT), reflect the range of signs and symptoms experienced by patients with PsA[2]
- Secukinumab is an interleukin (IL)-17A inhibitor that is efficacious across the updated GRAPPA-OMERACT PsA core domains[3,4]
 - However, the relative time to response across disease domains has not been studied

OBJECTIVE

- To evaluate time to achievement of clinical responses across the GRAPPA-OMERACT core domains in patients with PsA treated with secukinumab in the FUTURE 2-5 studies

METHODS

- This post hoc analysis evaluated data pooled from 1366 patients with PsA receiving secukinumab 300 or 150 mg in the phase 3 FUTURE 2-5 studies (NCT01752634, NCT01989468, NCT02294227, and NCT02404350)
 - Patients were randomized to secukinumab 300 mg every 4 weeks with loading dose (secukinumab at Weeks 0, 1, 2, 3, and 4, and every 4 weeks thereafter), secukinumab 150 mg every 4 weeks with or without loading dose, or placebo
 - Only patients randomized to secukinumab were included in this analysis; the 150-mg secukinumab treatment arms were combined for this analysis
- Efficacy outcomes assessed across GRAPPA-OMERACT PsA core domains included musculoskeletal disease activity, skin and nail disease activity, systemic inflammation, and patient-reported outcomes (**Figure 1**)
 - For each outcome, the proportion of patients achieving minimal clinically important difference (MCID) or complete resolution was assessed through Week 52 using as-observed data
 - Median time to the initial efficacy response and the response rate by Week 52 were estimated using the Kaplan-Meier method
 - No imputations were made for missing data

Figure 1. Outcomes assessed for median time to initial efficacy response and response rate by Week 52

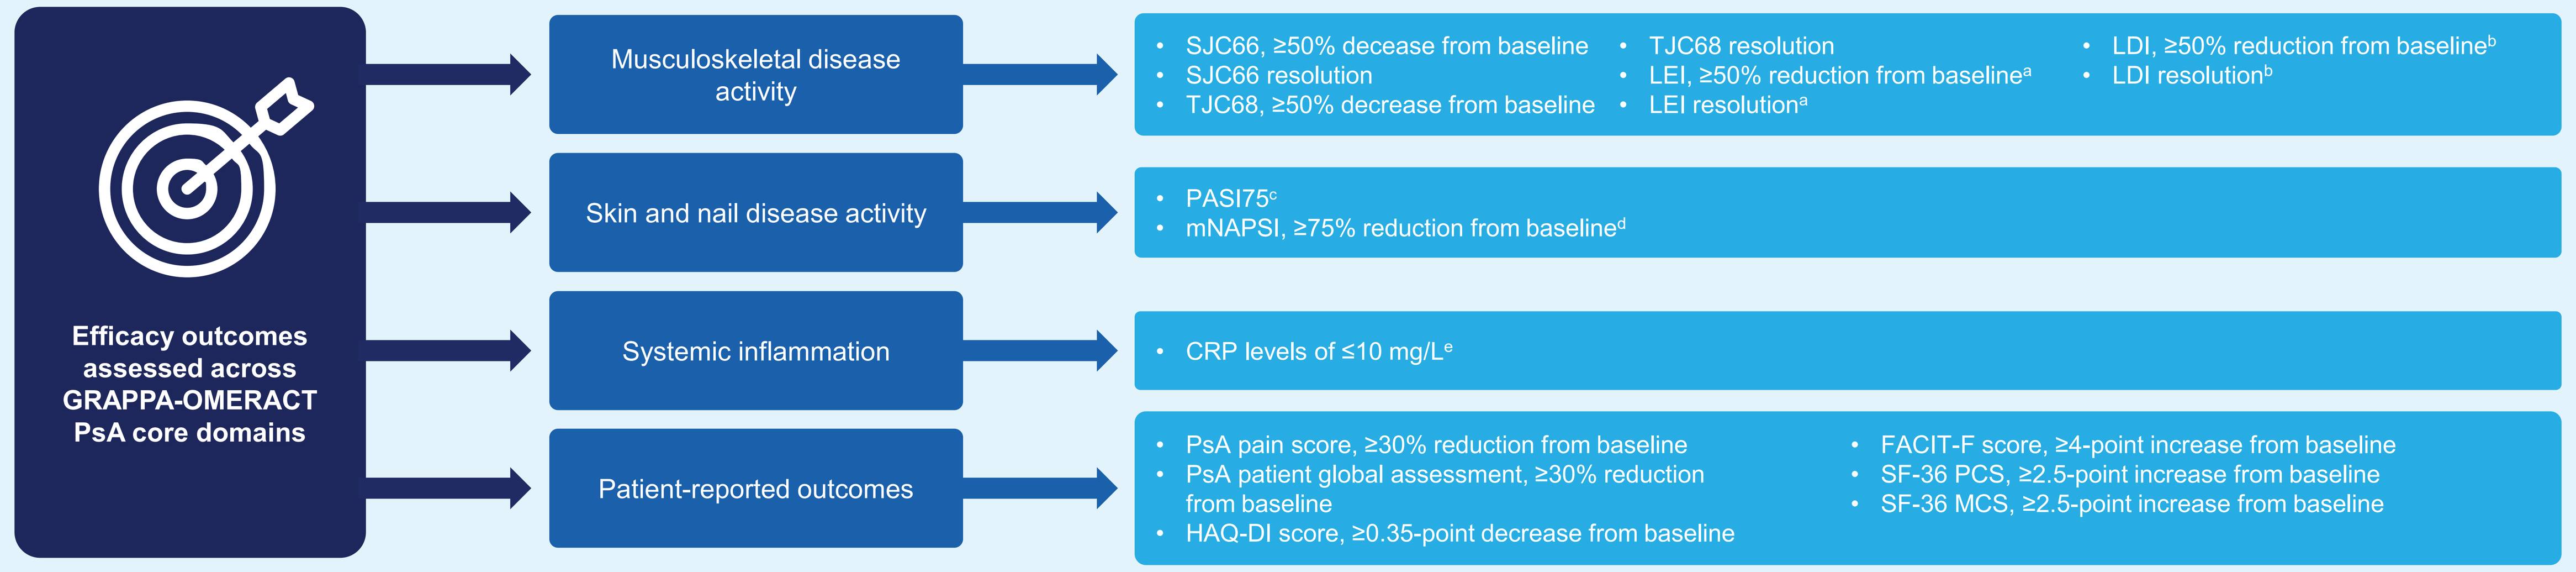
Efficacy outcomes assessed across GRAPPA-OMERACT PsA core domains
Musculoskeletal disease activity
• SJC66, ≥50% decease from baseline
• SJC66 resolution
• TJC68, ≥50% decrease from baseline
• TJC68 resolution
• LEI, ≥50% reduction from baseline[a]
• LEI resolution[a]
• LDI, ≥50% reduction from baseline[b]
• LDI resolution[b]
Skin and nail disease activity
• PASI75[c]
• mNAPSI, ≥75% reduction from baseline[d]
Systemic inflammation
• CRP levels of ≤10 mg/L[e]
Patient-reported outcomes
• PsA pain score, ≥30% reduction from baseline
• PsA patient global assessment, ≥30% reduction from baseline
• HAQ-DI score, ≥0.35-point decrease from baseline
• FACIT-F score, ≥4-point increase from baseline
• SF-36 PCS, ≥2.5-point increase from baseline
• SF-36 MCS, ≥2.5-point increase from baseline

CRP, C-reactive protein; FACIT-F, Functional Assessment of Chronic Illness Therapy — Fatigue; GRAPPA-OMERACT, Group for Research and Assessment of Psoriasis and Psoriatic Arthritis and Outcome Measures in Rheumatology; HAQ-DI, Health Assessment Questionnaire Disability Index; LDI, Leeds Dactylitis Index; LEI, Leeds Enthesitis Index; MCS, mental component summary; mNAPSI, modified Nail Psoriasis Severity Index; PCS, physical component summary; PASI75, Psoriasis Area and Severity Index, ≥75% reduction from baseline; PsA, psoriatic arthritis; SF-36, 36-Item Short Form Health Survey; SJC66, swollen joint count of 66 joints; TJC68, tender joint count of 68 joints.

[a] Among patients with enthesitis at baseline. [b] Among patients with dactylitis at baseline. [c] Among patients with psoriasis at baseline. [d] Among patients with nail psoriasis at baseline. [e] Among patients with CRP >10 mg/L at baseline.

RESULTS

Musculoskeletal disease activity, skin disease activity, and systemic inflammation

- Patients treated with either dose of secukinumab generally experienced rapid improvements across GRAPPA-OMERACT domains
- Achievement of MCID across musculoskeletal disease activity outcomes generally occurred within the first 4 weeks of secukinumab treatment (**Table 1**)
 - Across treatment arms, median time to achievement of MCID for musculoskeletal disease activity outcomes was generally similar; however, median time to achievement of a ≥50% decrease from baseline in tender joint count in 68 joints (TJC68) was shorter for patients receiving secukinumab 300 mg than for those receiving secukinumab 150 mg (approximately 4 vs 8 weeks)
- Median time to resolution of swollen joint count in 66 joints (SJC66), TJC68 (300-mg dose only), enthesitis, and dactylitis was approximately 18 to 24 weeks, 51 weeks, 8 to 12 weeks, and 4 weeks, respectively (**Table 1** and **Figure 2**)
 - Median time to resolution of SJC66 and enthesitis was shorter for patients receiving secukinumab 300 mg than for patients receiving secukinumab 150 mg
- Among patients with psoriasis at baseline, the median time to achievement of 75% reduction in the Psoriasis Area and Severity Index score (PASI75) was approximately 8 and 12 weeks for patients receiving secukinumab 300 mg and 150 mg, respectively (**Table 1**)
- Across treatment arms, patients with nail psoriasis at baseline achieved 75% improvement in the modified Nail Psoriasis Severity Index score (mNAPSI) after approximately 24 weeks of treatment (**Table 1**)
- Among patients with C-reactive protein (CRP) levels of >10 mg/L at baseline, median time to achievement of CRP levels of ≤10 mg/L was <2 weeks across treatment arms (**Figure 3** and **Table 1**)

Patient-reported outcomes

- Patients experienced rapid improvements in physical function (**Table 2**), pain (**Figure 4** and **Table 2**), and health-related quality of life (HRQOL) (**Figure 5** and **Table 2**) domains, consistent with the timing of improvements in musculoskeletal disease activity outcomes (approximately 4 weeks)
- Median achievement of MCID in the fatigue domain occurred slightly later, after approximately 8 weeks of secukinumab treatment (**Table 2**)
- Median time to achievement of MCID in pain, physical function, HRQOL, and fatigue domains was similar for patients receiving either secukinumab dose (**Table 2**)

Table 1. Median time to initial achievement of MCID or complete resolution in musculoskeletal disease activity, skin disease activity, and systemic inflammation domains through Week 52 in the FUTURE 2-5 trials[a]

Outcome	Secukinumab 300 mg (n=459)		Secukinumab 150 mg (n=907)	
	Time to response, median (95% CI), days	Achievement of initial response by Week 52, n/m [RR] (95% CI)	Time to response, median (95% CI), days	Achievement of initial response by Week 52, n/m [RR] (95% CI)
SJC66, ≥50% decrease from baseline	22 (NE-NE)	445/459 [98.7] (97.1-99.5)	27 (22-29)	866/907 [97.1] (95.8-98.1)
SJC66 resolution	127 (113-141)	350/459 [79.2] (75.2-82.9)	169 (143-177)	645/907 [75.1] (72.1-78.1)
TJC68, ≥50% decrease from baseline	29 (23-30)	425/459 [94.0] (91.5-96.0)	57 (36-57)	825/907 [93.9] (92.1-95.5)
TJC68 resolution	355 (281-NE)	231/459 [52.9] (48.2-57.7)	NE	386/907 [45.4] (42.0-48.8)
LEI, ≥50% reduction from baseline[b]	29 (22-29)	268/283 [95.8] (92.9-97.8)	29 (29-30)	512/569 [92.4] (89.9-94.5)
LEI resolution[b]	57 (32-58)	239/283 [86.4] (81.9-90.2)	85 (59-86)	442/569 [81.3] (77.8-84.6)
LDI, ≥50% reduction from baseline[c]	15 (15-16)	170/172 [99.3] (96.6-99.9)	15 (15-22)	314/329 [96.7] (94.3-98.3)
LDI resolution[c]	23 (16-29)	165/172 [97.5] (93.9-99.2)	29 (28-50)	302/329 [94.0] (90.9-96.4)
PASI75[d]	57 (57-59)	191/210 [92.5] (88.2-95.6)	86 (85-111)	373/476 [81.4] (77.5-84.9)
mNAPSI, ≥75% reduction from baseline[e]	164 (113-169)	212/272 [79.7] (74.6-84.4)	169 (169-171)	415/568 [76.1] (72.4-79.7)
CRP, ≤10 mg/L[f]	8 (NE-NE)	107/115 [94.0] (88.2-97.5)	12 (8-15)	188/219 [87.5] (82.5-91.6)

CRP, C-reactive protein; LDI, Leeds Dactylitis Index; LEI, Leeds Enthesitis Index; MCID, minimal clinically important difference; mNAPSI, modified Nail Psoriasis Severity Index; NE, not-estimable; n/m, number of patients who achieved initial response by Week 52/number of patients with corresponding baseline and post baseline assessments (as observed); PASI75, Psoriasis Area and Severity Index, ≥75% reduction from baseline; RR, response rate; SJC66, swollen joint count of 66 joints; TJC68, tender joint count of 68 joints.

[a] Median time and RR and associated 95% CIs are from the Kaplan-Meier estimate. [b] Among patients with enthesitis at baseline. [c] Among patients with dactylitis at baseline. [d] Among patients with psoriasis at baseline. [e] Among patients with nail psoriasis at baseline. [f] Among patients with CRP >10 mg/L at baseline.

Table 2. Median time to initial achievement of MCID in patient-reported outcomes related to pain, HRQOL, fatigue, and physical function domains through Week 52 in the FUTURE 2-5 trials[a]

Outcome	Secukinumab 300 mg (n=459)		Secukinumab 150 mg (n=907)	
	Time to response, median (95% CI), days	Achievement of initial response by Week 52, n/m [RR] (95% CI)	Time to response, median (95% CI), days	Achievement of initial response by Week 52, n/m [RR] (95% CI)
PsA pain score, ≥30% reduction from baseline	22 (22-23)	400/459 [88.8] (85.5-91.6)	23 (22-29)	775/905 [87.8] (85.4-89.9)
PsA patient global assessment, ≥30% reduction from baseline	22 (22-25)	409/459 [90.2] (87.2-92.8)	26 (22-29)	764/905 [86.8] (84.4-89.0)
HAQ-DI score, ≥0.35-point decrease from baseline	22 (22-25)	352/457 [77.7] (73.8-81.5)	29 (22-30)	658/904 [74.0] (71.0-76.9)
FACIT-F score, ≥4-point increase from baseline	52 (32-57)	357/452 [80.4] (76.5-84.1)	57 (37-57)	692/891 [79.3] (76.5-82.0)
SF-36 PCS, ≥2.5-point increase from baseline	29 (NE-NE)	408/454 [90.8] (87.8-93.3)	29 (22-29)	765/898 [86.3] (83.9-88.6)
SF-36 MCS, ≥2.5-point increase from baseline	30 (29-33)	354/454 [78.9] (74.9-82.6)	29 (24-29)	717/898 [80.8] (78.1-83.3)

FACIT-F, Functional Assessment of Chronic Illness Therapy — Fatigue; HAQ-DI, Health Assessment Questionnaire Disability Index; HRQOL, heath-related quality of life; MCID, minimal clinically important difference; MCS, mental component summary; n/m, number of patients who achieved initial response by Week 52/number of patients with corresponding baseline and post baseline assessments (as observed); PCS, physical component summary; PsA, psoriatic arthritis; RR, response rate; SF-36, 36-Item Short Form Health Survey.

[a] Median time and RR and associated 95% CIs are from the Kaplan-Meier estimate.

Figure 2. Kaplan-Meier time curve showing achievement of LDI resolution[a] through Week 52

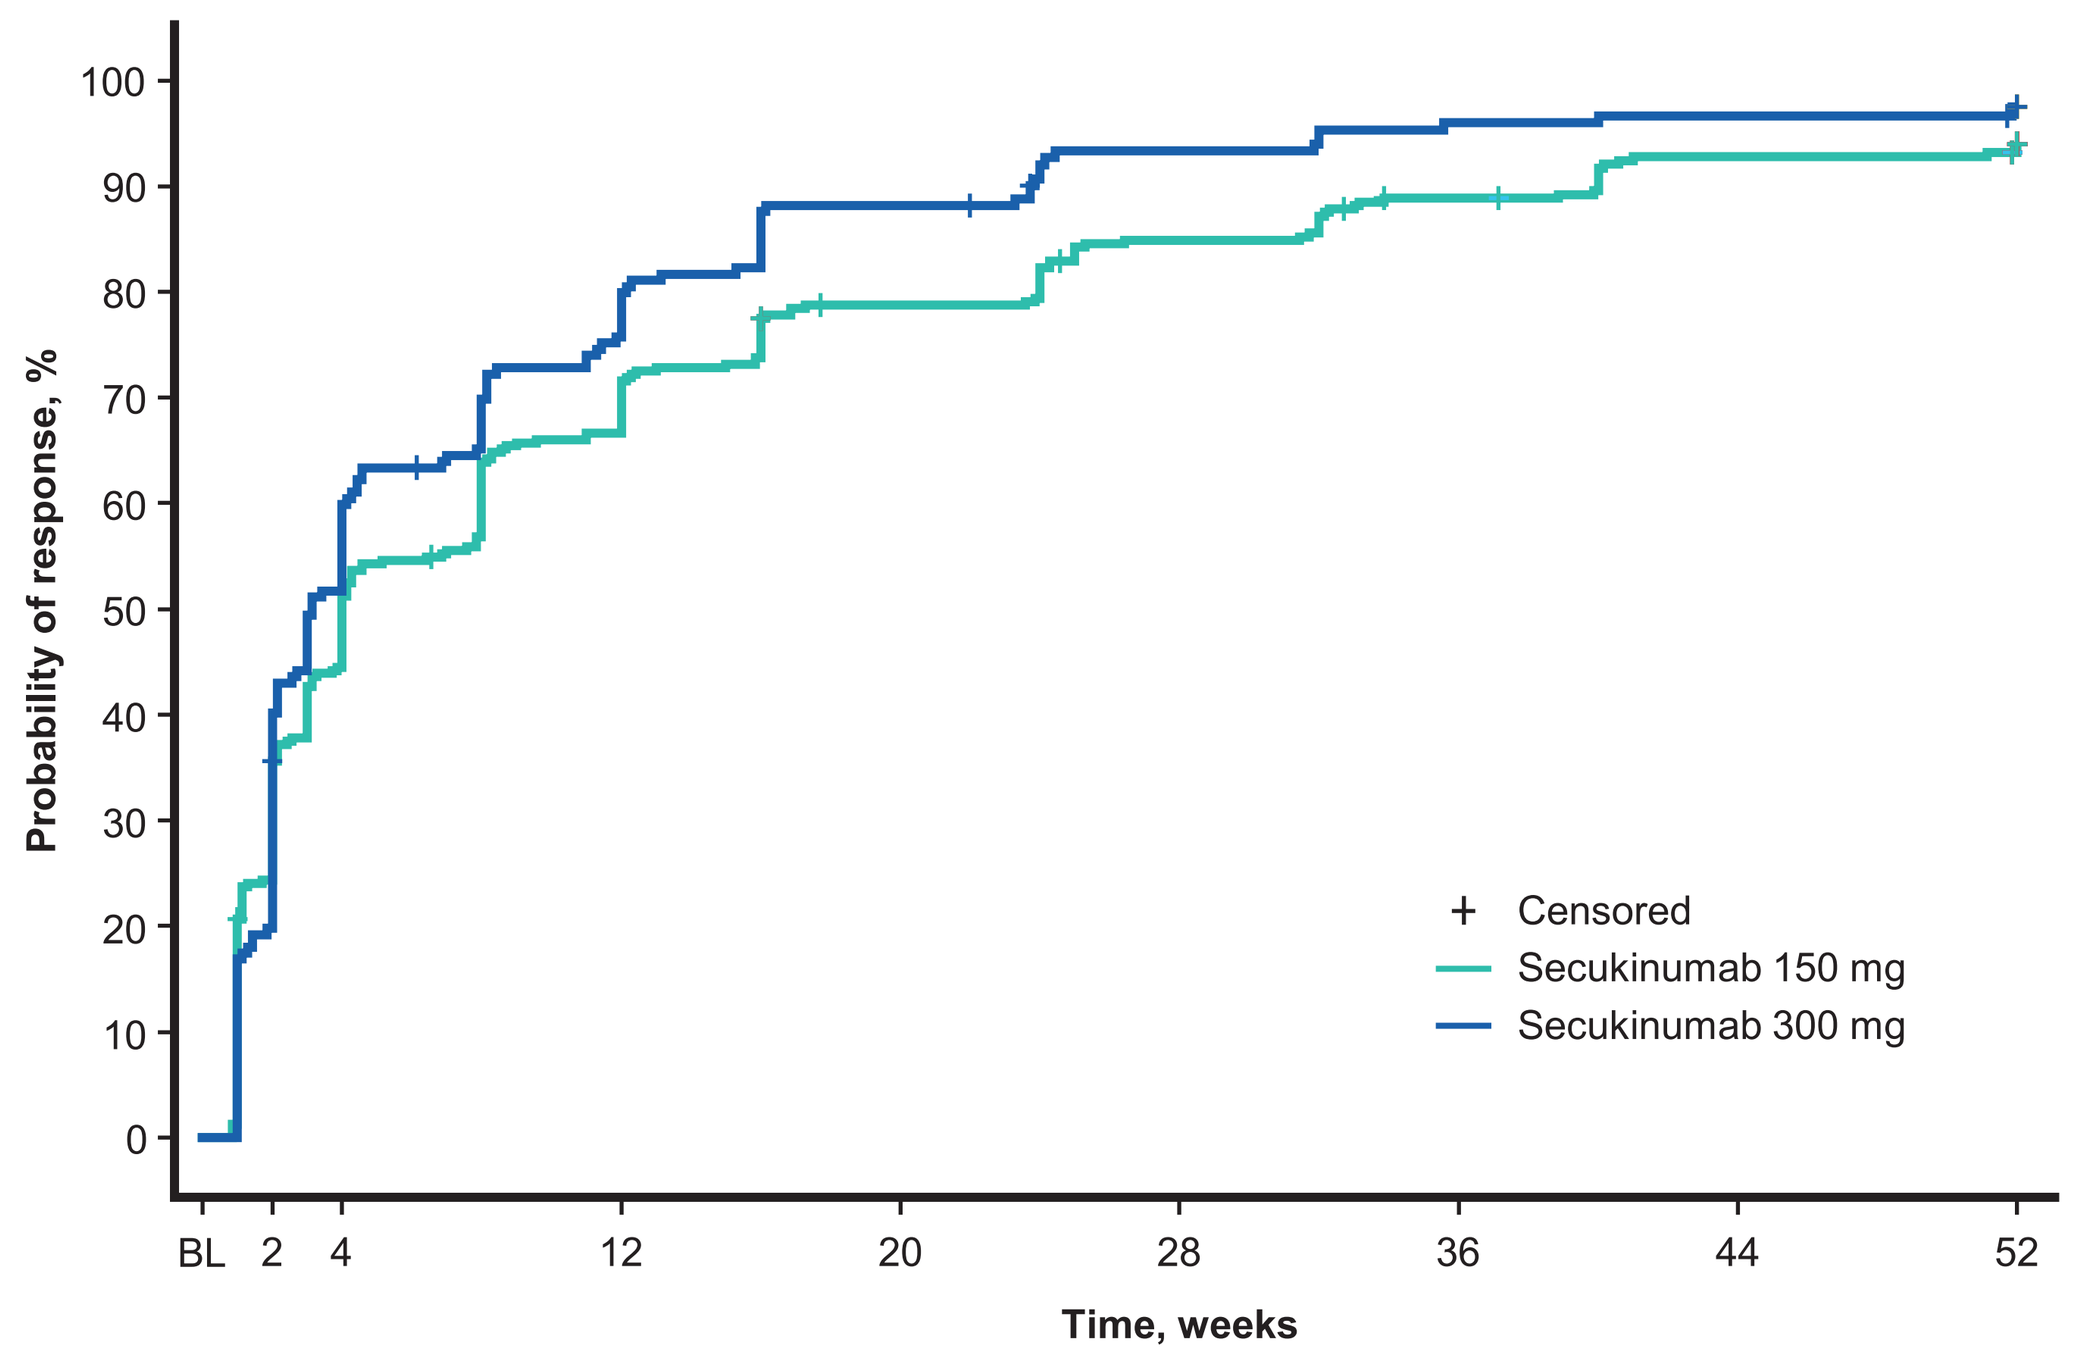
Probability of response, %
Time, weeks
+ Censored
Secukinumab 150 mg
Secukinumab 300 mg

No. at risk/events

	BL	1	2	3	4	8	12	16	20	24	28	32	36	40	44	48	52
Secukinumab 150 mg	329/0	325/68	248/117	203/140	181/168	140/209	108/234	85/253	66/257	64/268	46/276	44/283	32/288	29/296	20/299	20/299	16/302
Secukinumab 300 mg	172/0	172/29	138/69	96/85	83/103	59/120	41/137	30/150	20/151	14/157	10/159	9/162	6/163	6/164	5/164	5/164	3/165

BL, baseline; LDI, Leeds Dactylitis Index.

[a] Among patients with dactylitis at baseline.

Figure 3. Kaplan-Meier time curve showing achievement of ≤10 mg/L CRP levels[a] through Week 52

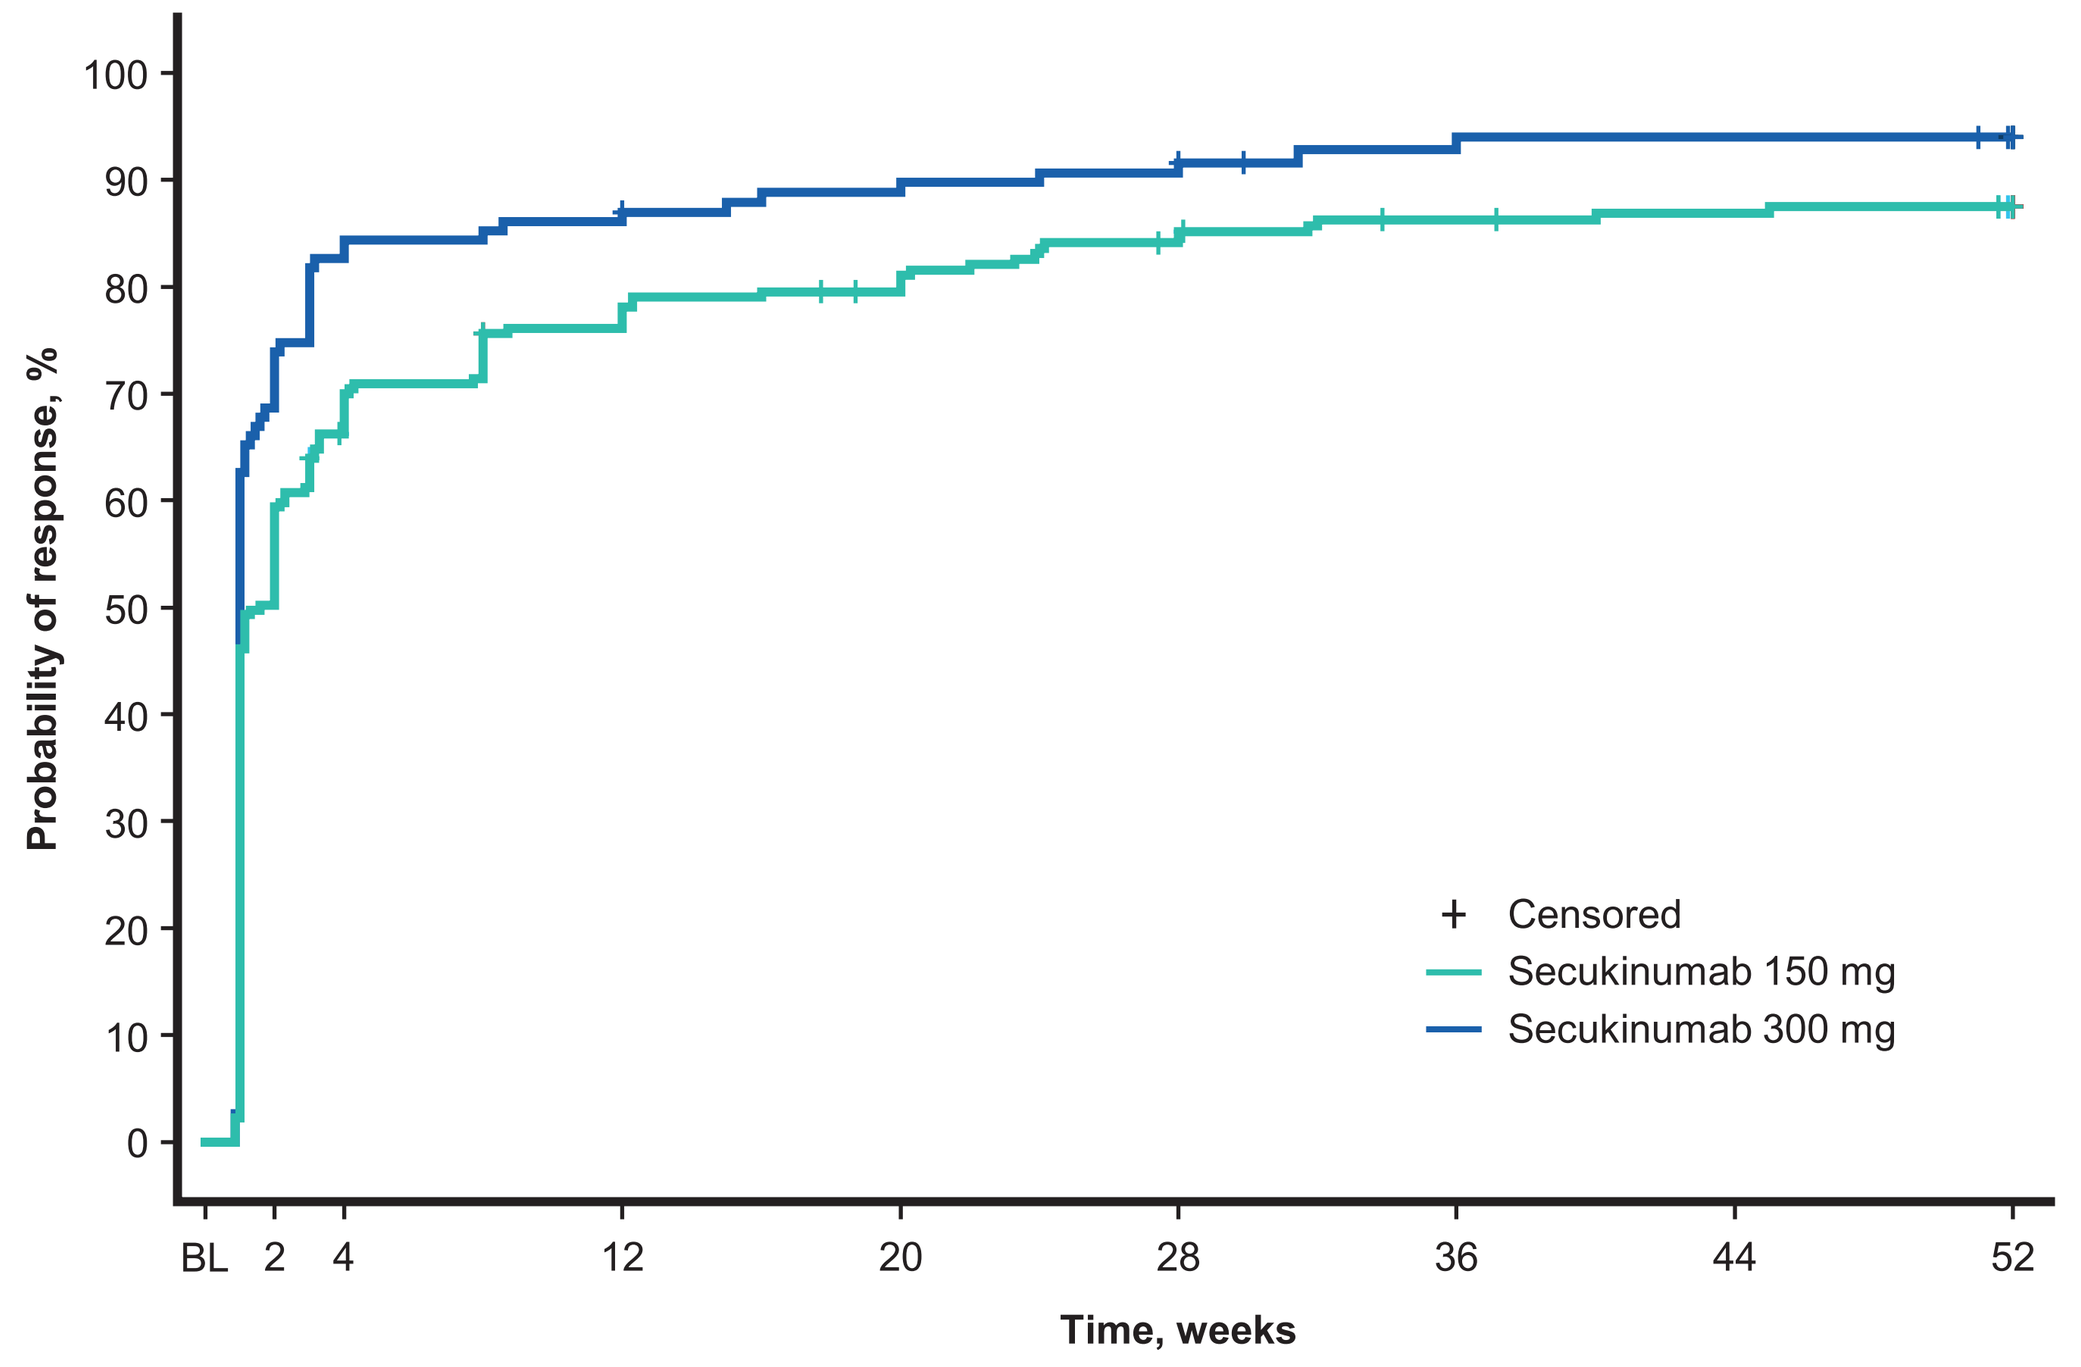
Probability of response, %
Time, weeks
+ Censored
Secukinumab 150 mg
Secukinumab 300 mg

No. at risk/events

	BL	1	2	3	4	8	12	16	20	24	28	32	36	40	44	48	52
Secukinumab 150 mg	219/0	214/101	109/130	85/140	72/153	61/165	49/170	43/173	40/176	33/181	30/184	26/186	24/186	23/187	22/187	21/188	18/188
Secukinumab 300 mg	115/0	112/72	36/85	29/94	20/97	18/98	16/100	13/102	12/103	11/104	10/105	6/106	6/107	5/107	5/107	5/107	3/107

BL, baseline; CRP, C-reactive protein.

[a] Among patients with CRP >10 mg/L at baseline.

Figure 4. Kaplan-Meier time curve showing achievement of a ≥30% reduction in PsA pain score from baseline through Week 52

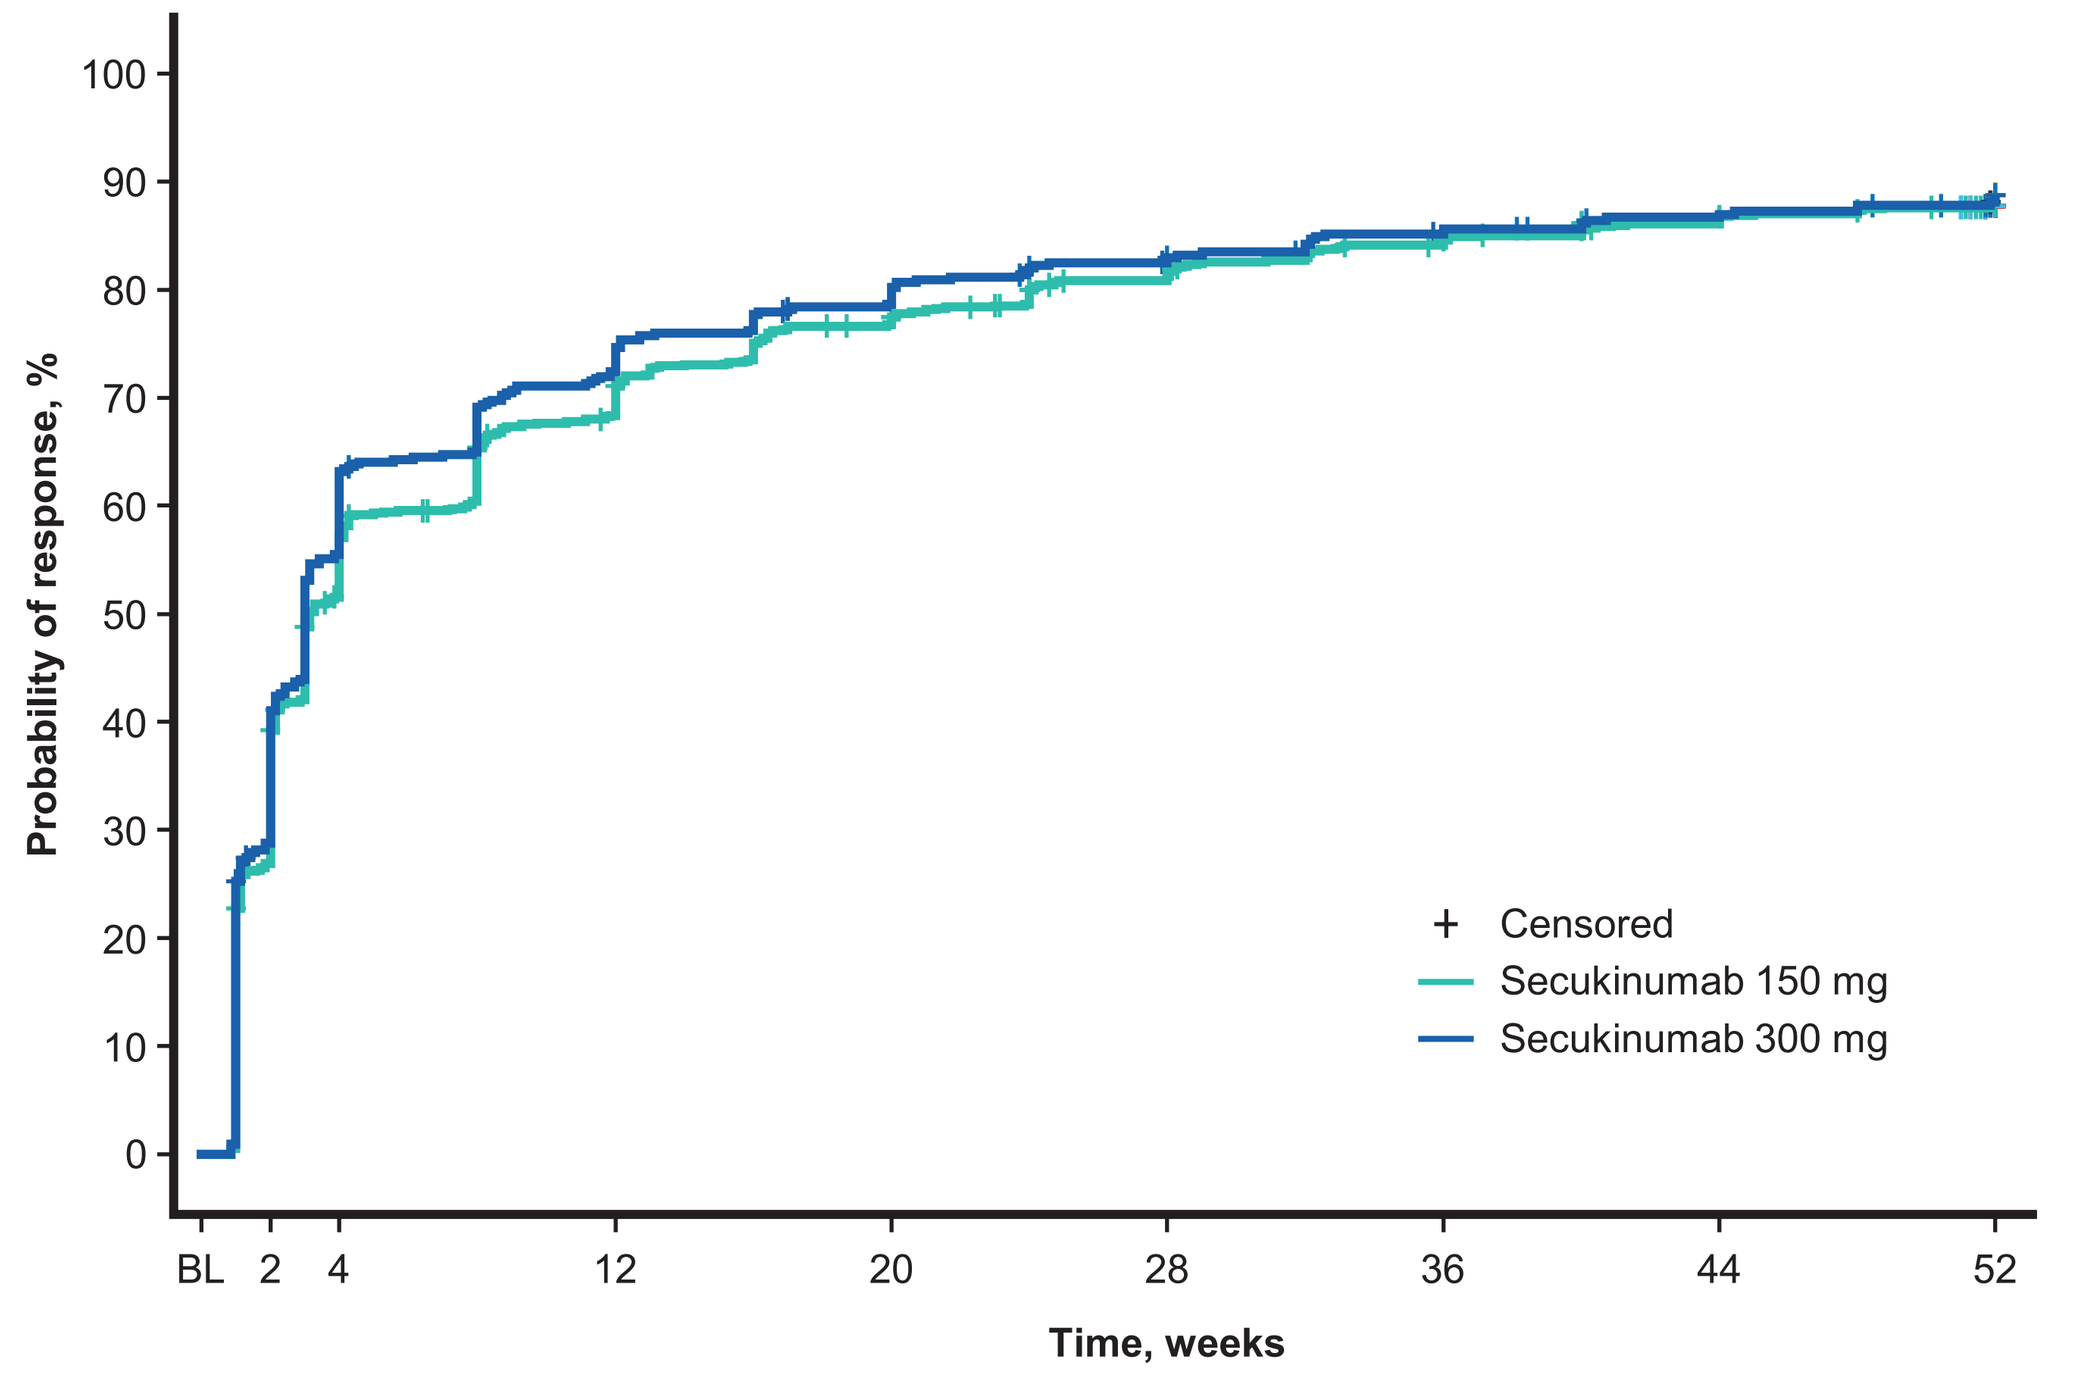
Probability of response, %
Time, weeks
+ Censored
Secukinumab 150 mg
Secukinumab 300 mg

No. at risk/events

	BL	1	2	3	4	8	12	16	20	24	28	32	36	40	44	48	52
Secukinumab 150 mg	905/0	900/206	661/355	522/441	433/515	351/589	276/639	230/673	200/694	178/715	154/729	136/742	122/752	112/759	102/768	94/772	70/775
Secukinumab 300 mg	459/0	455/116	325/188	256/243	203/289	159/316	125/341	108/355	94/366	80/374	74/378	68/383	60/389	56/391	49/394	47/397	35/400

BL, baseline; PsA, psoriatic arthritis.

Figure 5. Kaplan-Meier time curve showing achievement of a ≥0.35-point decrease in HAQ-DI score from baseline through Week 52

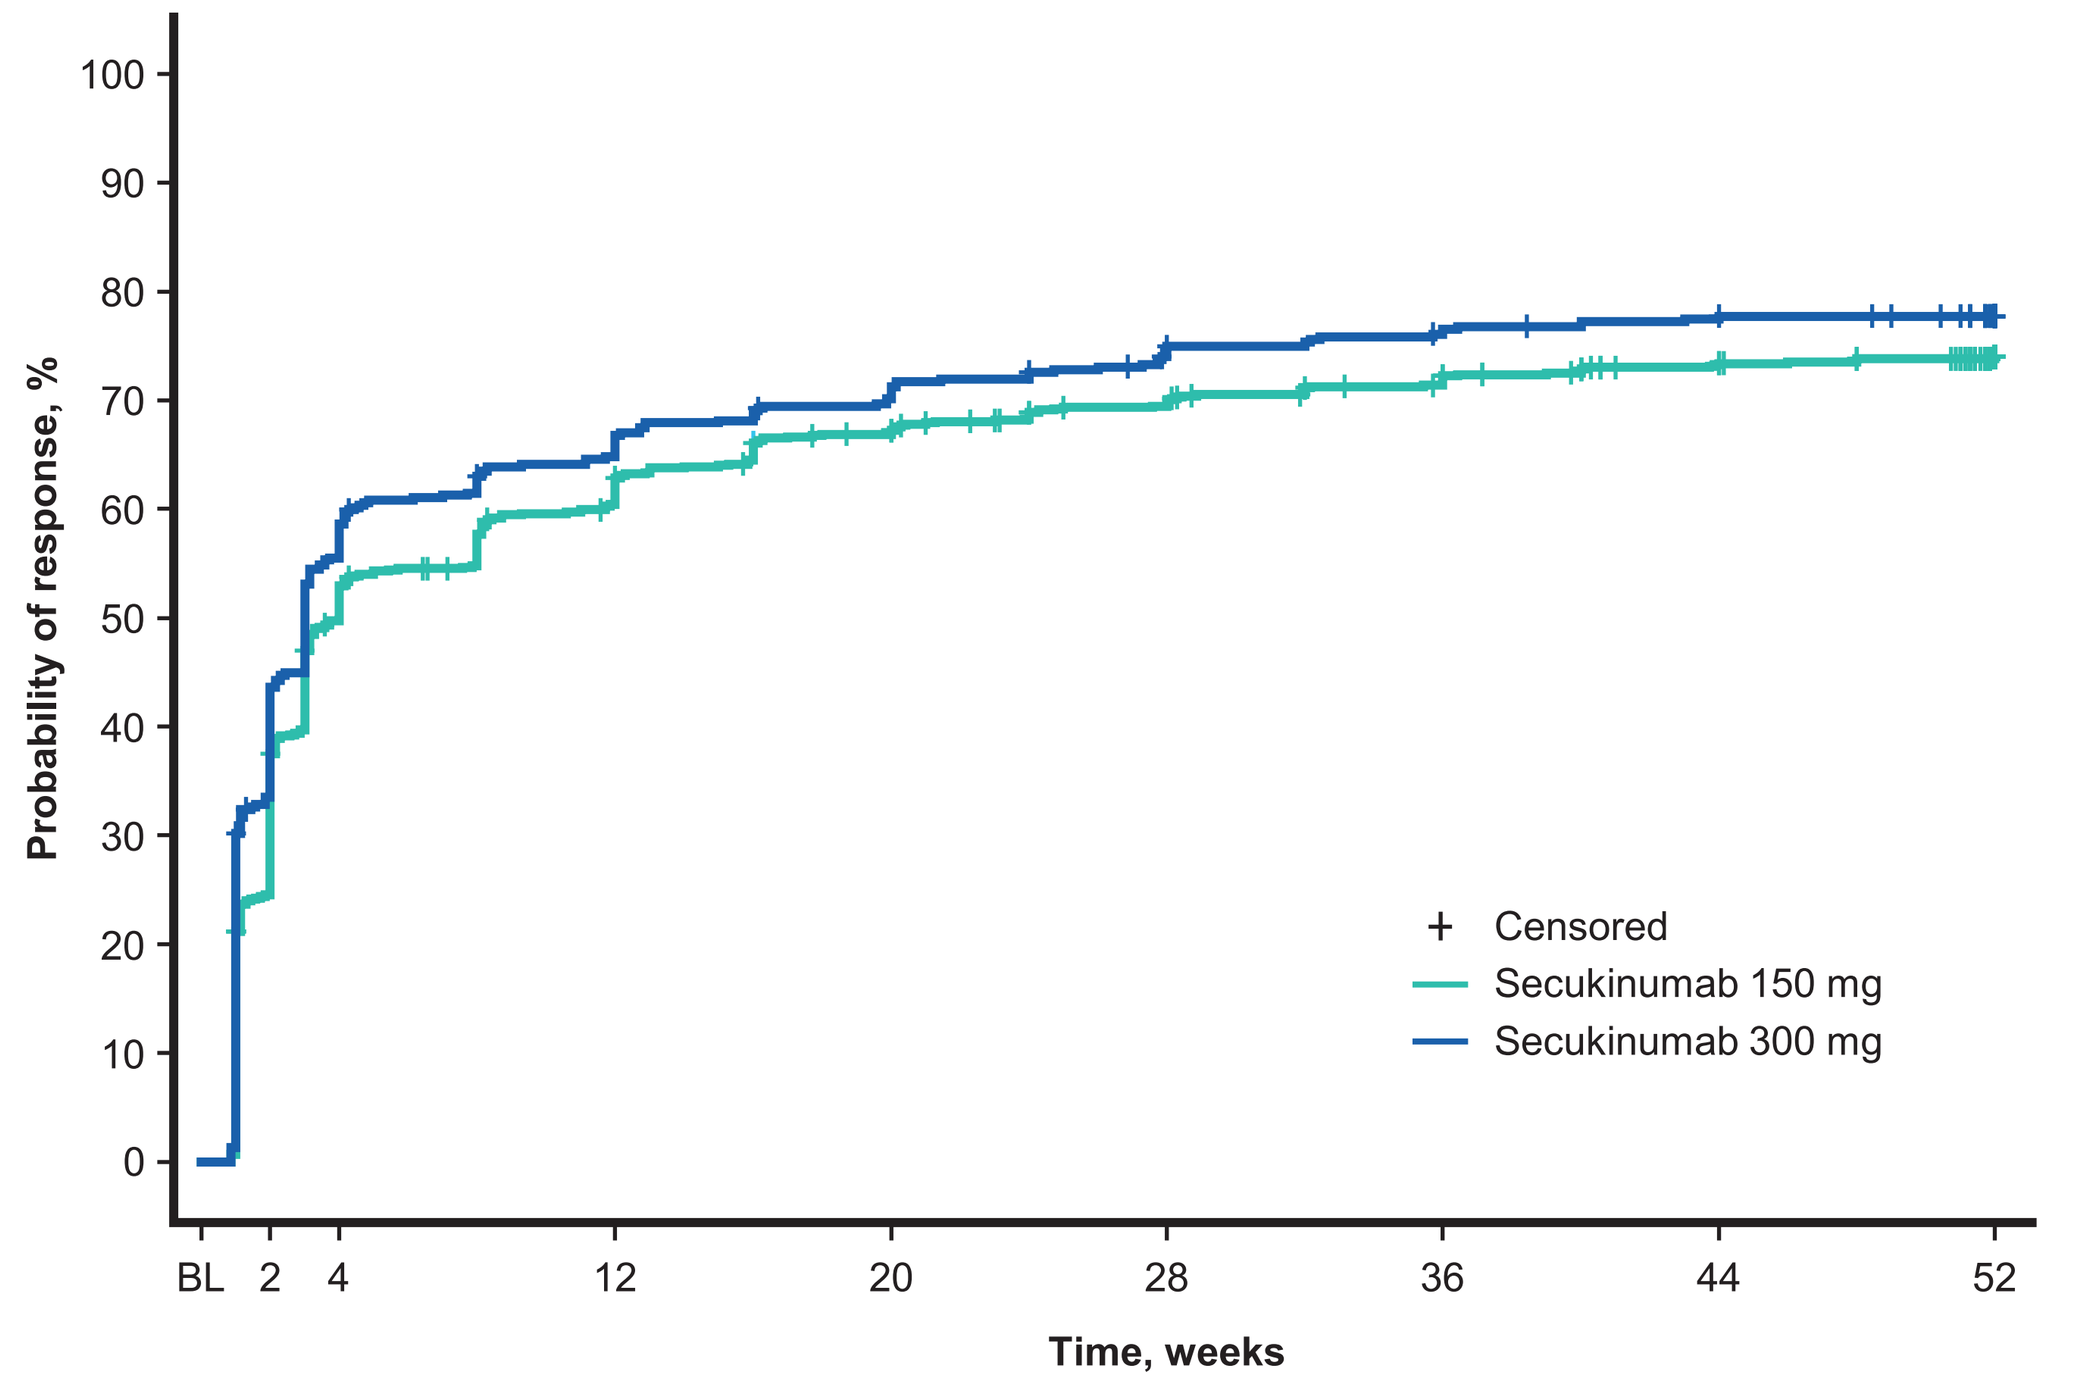
Probability of response, %
Time, weeks
+ Censored
Secukinumab 150 mg
Secukinumab 300 mg

No. at risk/events

	BL	1	2	3	4	8	12	16	20	24	28	32	36	40	44	48	52
Secukinumab 150 mg	904/0	898/191	679/338	542/424	450/477	401/519	349/565	311/593	287/603	268/617	255/627	242/636	232/645	217/650	204/654	199/657	163/658
Secukinumab 300 mg	457/0	451/138	302/199	250/242	202/267	174/287	158/304	143/314	133/324	125/330	113/340	108/342	102/347	98/350	95/352	93/352	75/352

BL, baseline; HAQ-DI, Health Assessment Questionnaire Disability Index.

References

1. Ritchlin CT, et al. *N Engl J Med*. 2017;376(21):2095-2096.
2. Orbai AM, et al. *Ann Rheum Dis*. 2017;76(4):673-680.
3. Orbai AM, et al. *J Rheumatol*. 2020;47(6):854-864.
4. Orbai AM, et al. *Rheumatol Ther*. 2021;8(3):1223-1240.

Acknowledgments

This study was sponsored by Novartis Pharmaceuticals Corporation, East Hanover, NJ. The funding source was involved in study design, data analysis, drafting, and approval of this poster. Medical writing support was provided by Katherine West, PhD, of Nucleus Global, an Inizio Company, and funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP 2022) guidelines. Authors had full control of the content and made the final decision on all aspects of this publication.

Disclosures

L.C. Coates has received grants/research support from AbbVie, Amgen, Celgene, Eli Lilly, Janssen, Novartis, Pfizer, and UCB; has worked as a paid consultant for AbbVie, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Eli Lilly, Gilead, Galapagos, Janssen, MoonLake, Novartis, Pfizer, and UCB; and has been paid as a speaker for AbbVie, Amgen, Biogen, Celgene, Eli Lilly, Galapagos, Gilead, GSK, Janssen, Medac, Novartis, Pfizer, and UCB. **I. McInnes** received honoraria from AbbVie, AstraZenaca, BMS, Boehringer, Compugen, Cabaletta, Causeway, Dextera, Eli Lilly, MoonLake, Pfizer, Novartis, Janssen, and UCB; and research support from Versus Arthritis, MRC & Wellcome Trust. **E. Husni** has consulted for AbbVie, Bristol Myers Squibb, Eli Lilly, Janssen, Novartis, Pfizer, and UCB. **C. Vizcaya** and **W. Bao** are employees of and own stock in Novartis. **P. Mease** has received research grants from AbbVie, Acelyrin, Amgen, Bristol Myers Squibb, Gilead, Janssen, Eli Lilly, Novartis, Pfizer, Sun Pharma, and UCB; consulting fees from AbbVie, Acelyrin, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Galapagos, Gilead, GSK, Janssen, Eli Lilly, MoonLake Pharma, Novartis, Pfizer, Sun Pharma, Takeda, UCB, and Ventyx; and speakers bureau fees from AbbVie, Amgen, Janssen, Eli Lilly, Novartis, Pfizer, and UCB.